ECONOMIC BURDEN OF SPINAL MUSCULAR ATROPHY IN ITALY

Andrea Marcellusi[1,2], Chiara Bini[1], Jacopo Casiraghi[3],
Francesco D'Ambrosio[1], Maria A Rotundo[1] , Anita Pallara[3], Francesco S Mennini[1,2] on behalf of fam sma Onlus Italy

[1]Economic Evaluation and HTA (EEHTA) - Faculty of Economics, University of Rome "Tor Vergata", Rome, Italy. [2]Institute of Leadership and Management in Health - Kingston University London, London, UK. [3]Famiglie SMA Onlus Italy

BACKGROUND

Spinal muscular atrophy (SMA) is an autosomal recessive, inherited neuromuscular disease that affects the spinal anterior nerve cells. It is characterized by atrophy and weakness of the skeletal muscles of the limbs and trunk of the bulbar and respiratory muscles. It is the most common genetic disease for infant mortality with an estimated incidence of 1/10.000 live births [1] and a prevalence rate 1.31-1.87/100.000 [2]. In Italy, an estimated 850 persons are living with SMA [3].
The classification system for SMA is based on the age at symptom onset and the most advanced motor milestone attained during development. The phenotypes in which it is classified are three[4]:

- type I, with onset <6 months, is the most severe subtype with the highest incidence and lowest prevalence. Patients never acquire the ability to sit unsupported;
- type II is intermediate type between type I and type III and its onset is between 7 and 18 months. Patients achieve the ability to sit unsupported and some of them are able to acquire standing position, but they do not acquire the ability to walk independently;
- type III, with onset >18 months represents the mildest one. They typically reach all major motor milestones, as well as independent walking. However during infancy they develop proximal muscular weakness.

The difficulties of living with SMA begin with the long and often arduous process of diagnosis and living with SMA is challenging not only for patients but also for their families and caregivers, as well as medical personnel and the society.
A study conducted in 2018 in Italy [5] showed that SMA imposes a considerable economic burden on society both in terms of direct costs (treatment, hospitalizations, emergency consultations, visits i.e. neurologist, psychologist…) and indirect costs (economic loss of productivity of patients and/or their caregivers due to absenteeism and presenteeism).
Nusinersen is a synthetic anti-sense oligonucleotide that modifies splicing of SMN2 precusor mRNA, there by increasing the levels of functional full-length SMN protein [6].

OBJECTIVES

The aim of this study was to estimate the indirect and direct costs associated with SMA in Italy in patients treated with nusinersen and compare the same results estimated for the not treated population of SMA in the 2018 study [5].

METHODS

In order to develop the economic model, the multidisciplinary group of researchers in charge of the study on the costs associated with SMA in 2018 [5], developed a new questionnaire in collaboration with *Famiglie SMA*.
Data collection started in February 2019 and ended in September 2019 and questionnaire has been compiled by patients treated or not treated with nusinersen and caregivers.
At the end of this period, all the data were analyzed within the economic model and have been estimated: direct health costs incurred by NHS (visits and clinical tests), direct health costs incurred by patient (visits, clinical tests, devices excluded public reimbursement and healthcare professionals), and indirect costs in terms of loss of productivity at work.
Below are presented results from a preliminary analysis.

RESULTS

Preliminary analysis considered 24 completed questionnaires (23 from caregivers and 1 from an adult patient). Of these, 67% (16 patients) were treated with nusinersen, 4% (1 patient) were suspended their treatment and 16% (4 patients) were had not treated with nusinersen; the remaining 3 patients (13%) did not responded. Of 24 respondents, 3 (13%) were patients with SMA I, 16 (67%) SMA II and 2 (8%) SMA III (3 did not respond).
SMA III was not selected in the cost calculation because we only had two patients. The average patients age was 8.53 years (average age at diagnosis 0.8 years) with a greater proportion of women (55%).
About 75% of patients had assistance from their family members, and 94% of them need support from a second caregiver.

From the caregiver perspective, 58.33% of them were employed and about 80% of them declared that they lost workdays due to the assistance during last year (41.43 days lost of workday on average).
The only adult patient participating in the study was employed and declared an average reduction in production capacity of about 38% (96 days lost per year on average).
Figure 1 show the indirect and direct costs for SMA type I (3 patients) and type II (16 patients); type III were excluded from this analysis because only 2 patients were treated in the sample.
Figure 2 compares the average costs for patient treated and untreated [5] considering only SMA type I and II.
Direct costs sustained by patients (e.g., visits, devices, healthcare professionals, etc.) for treated patients tend to be higher vs. untreated patients; this might due to an increasing in the monitoring visits and follow-up needed for the treatment.

Figure 1: Distribution of direct and indirect costs for SMA patients treated with nusinersen (Sample of 19 patients)

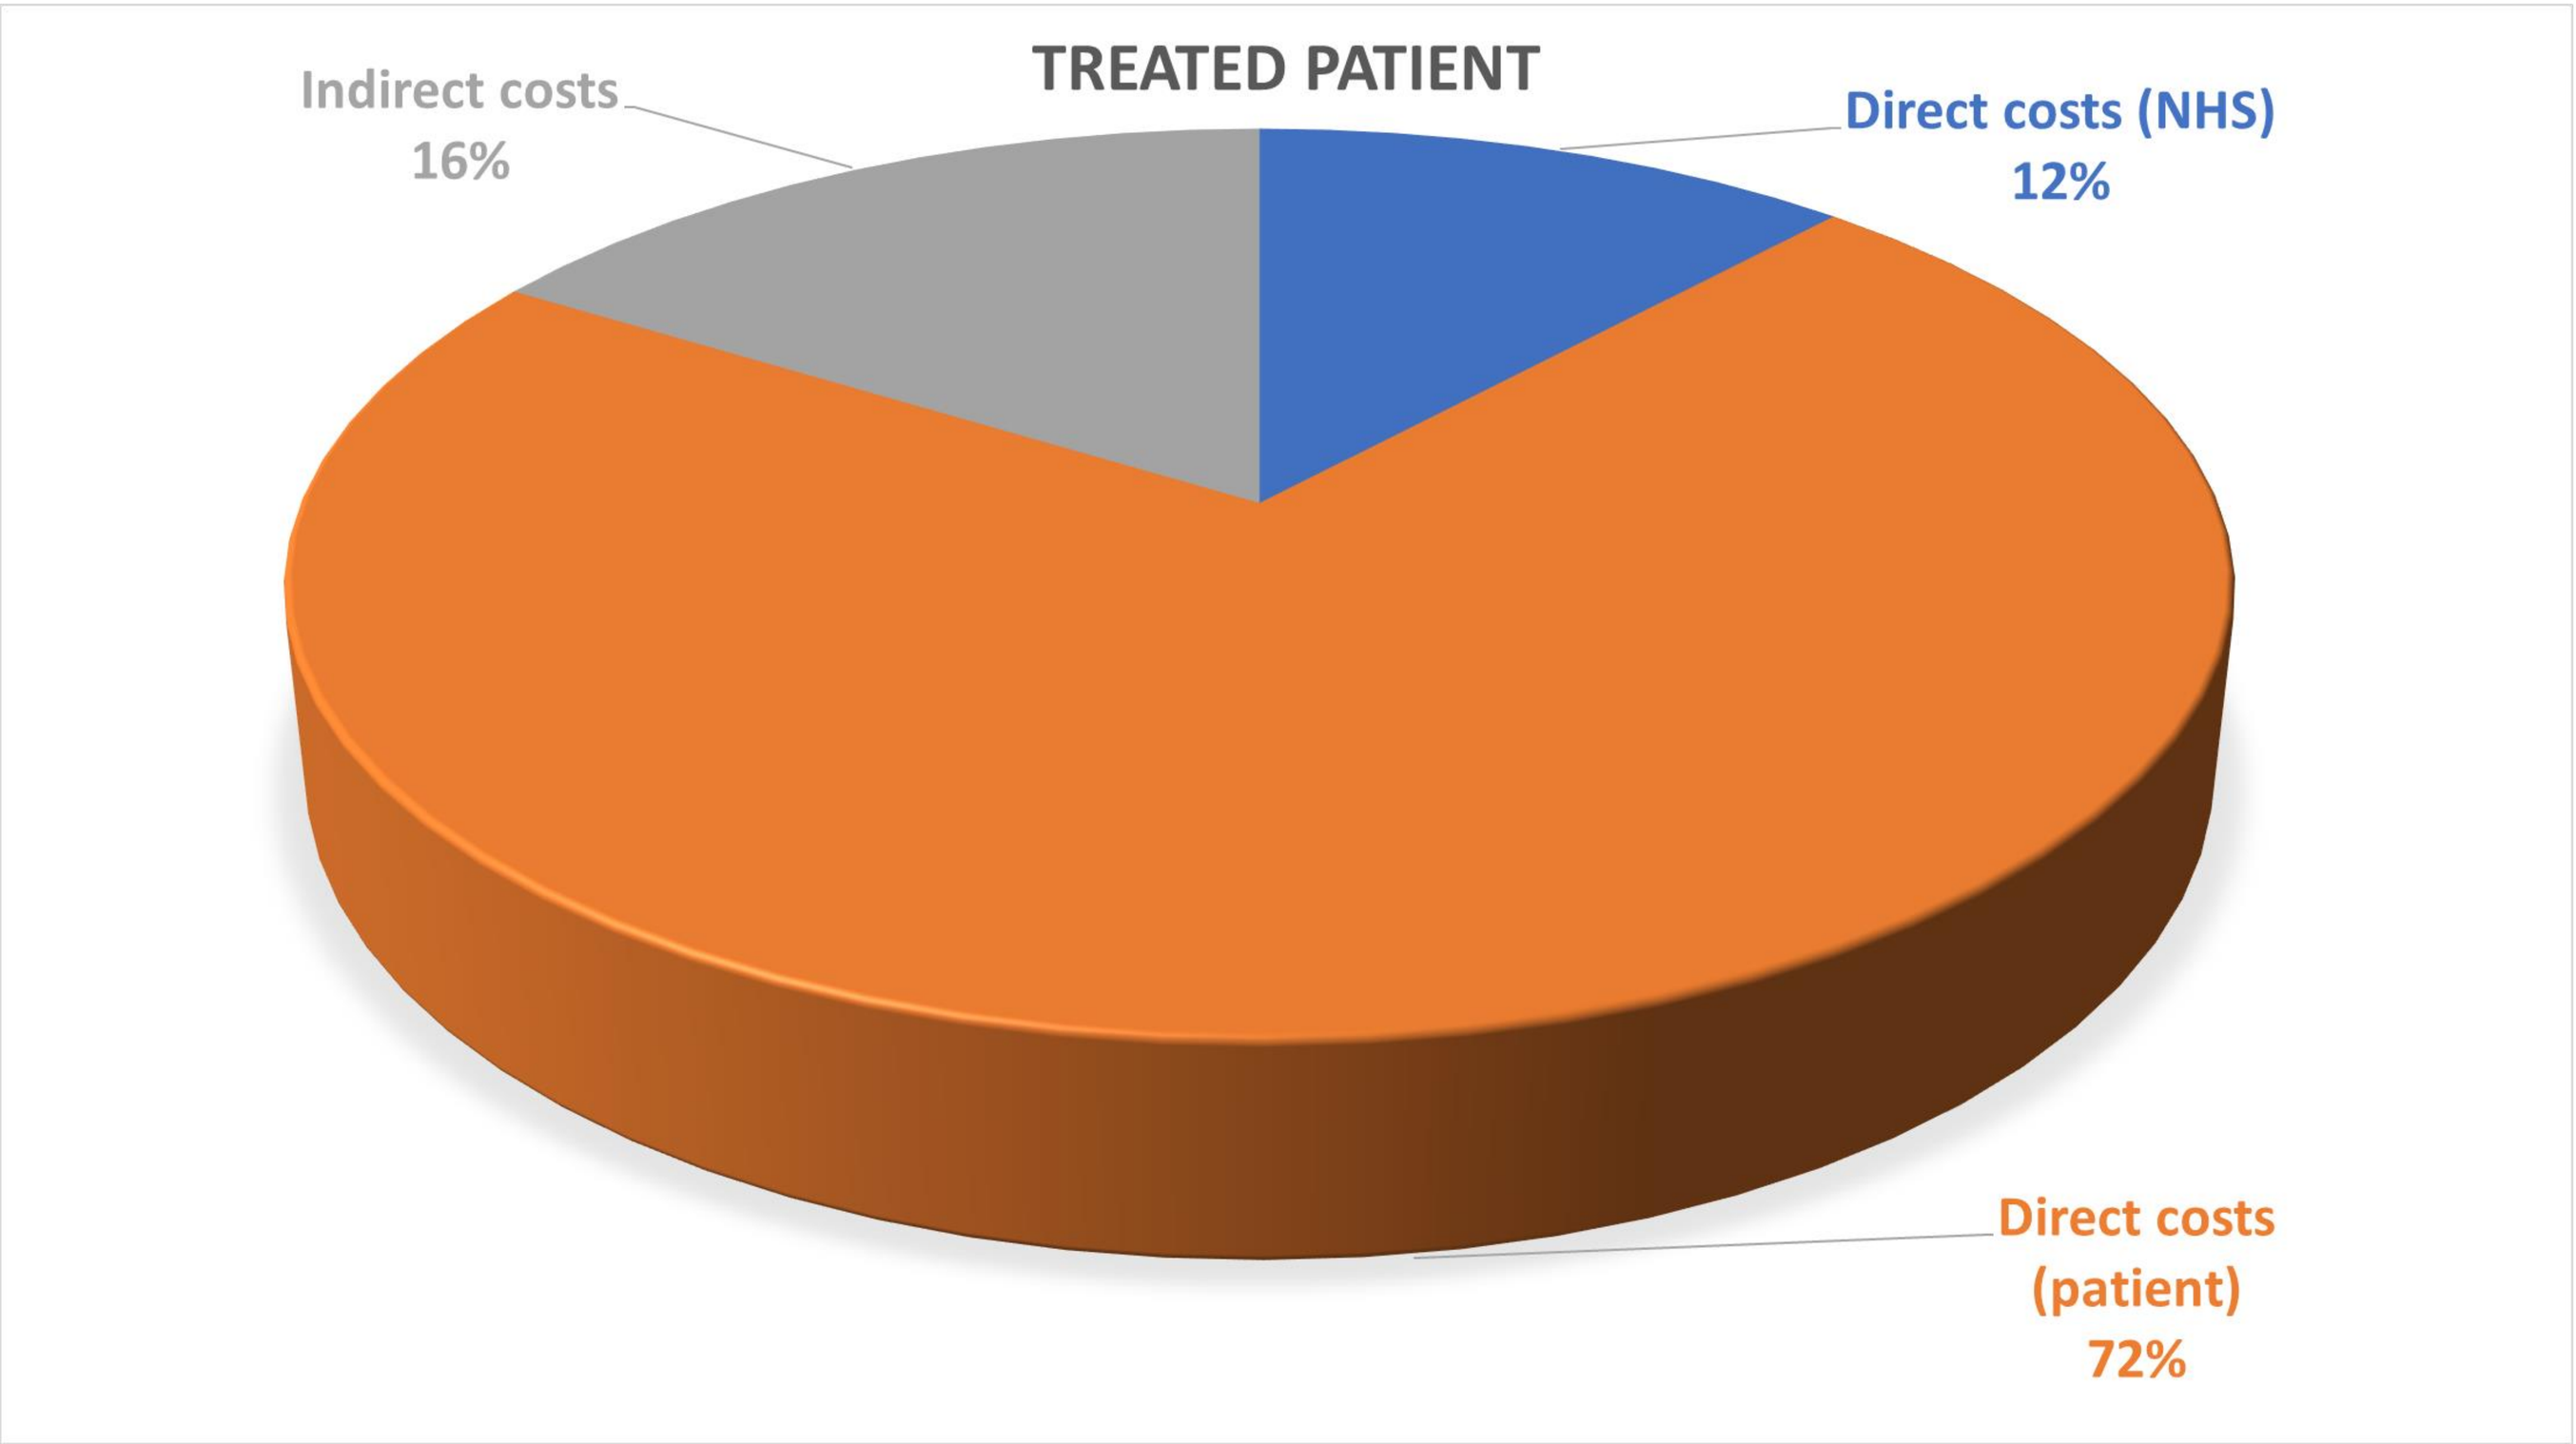
TREATED PATIENT
Indirect costs 16%
Direct costs (NHS) 12%
Direct costs (patient) 72%

Figure 2: Average cost per Treated (Sample of 19 patients) and Untreated (Sample of 118 patients) patient

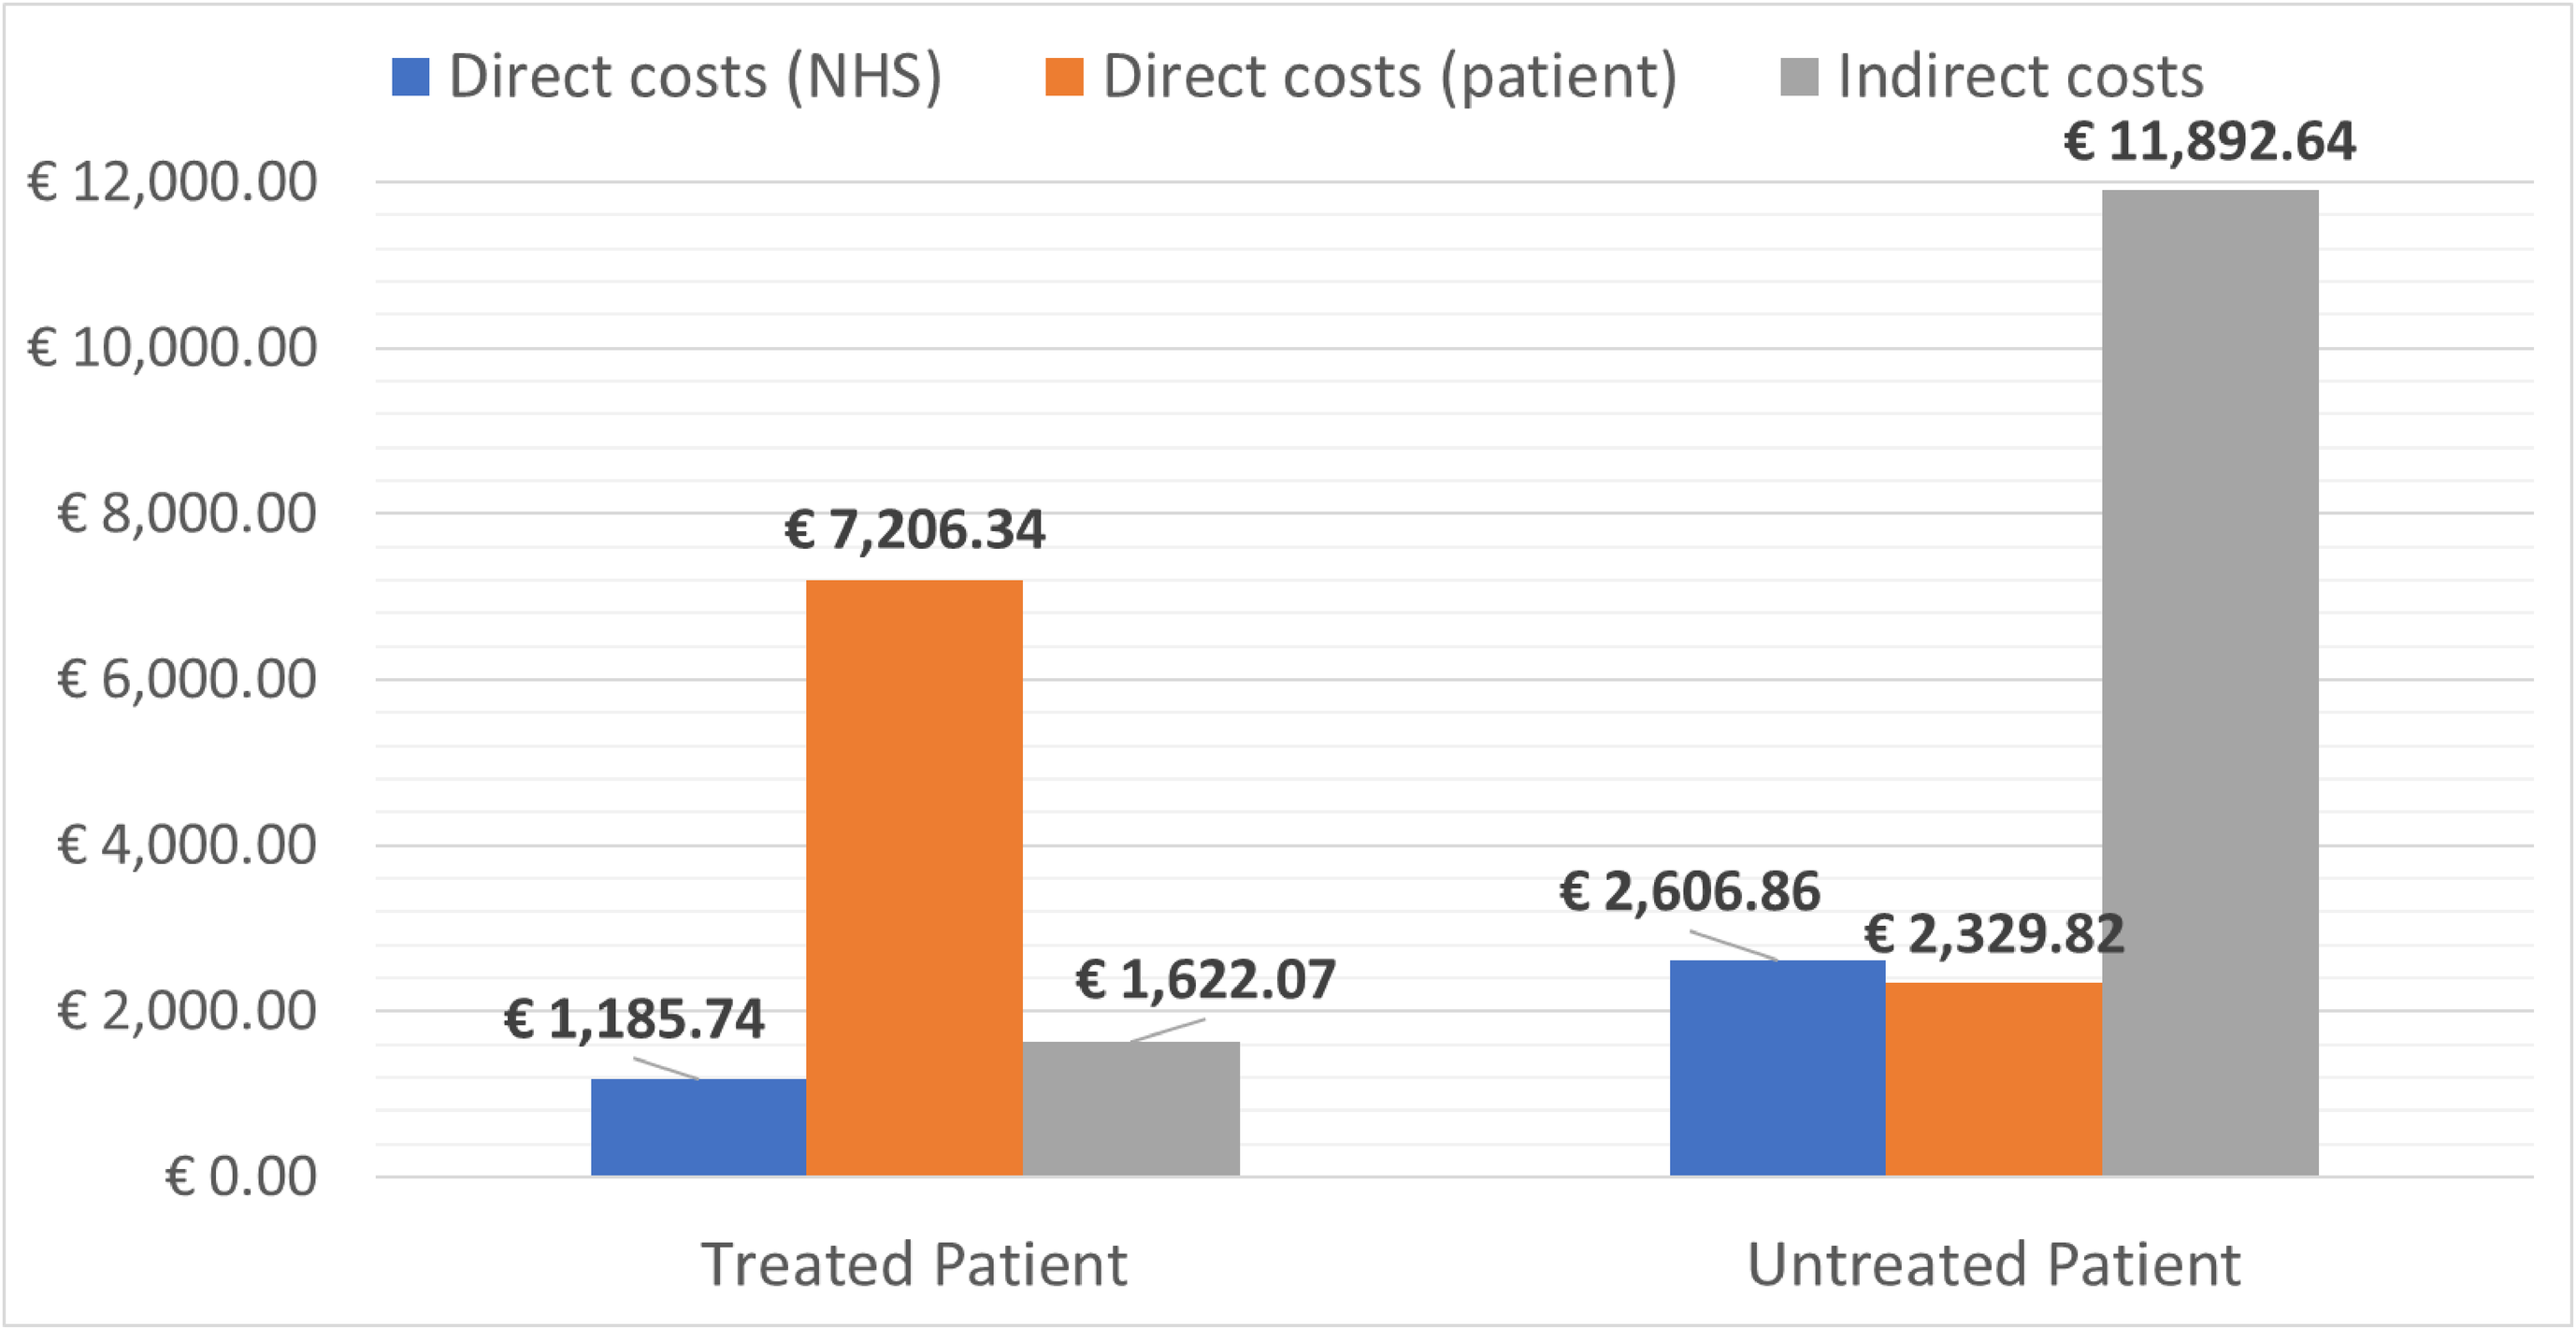
Direct costs (NHS)
Direct costs (patient)
Indirect costs
€ 12,000.00
€ 10,000.00
€ 8,000.00
€ 6,000.00
€ 4,000.00
€ 2,000.00
€ 0.00
€ 1,185.74
€ 7,206.34
€ 1,622.07
€ 2,606.86
€ 2,329.82
€ 11,892.64
Treated Patient
Untreated Patient

LIMITATIONS

- The results of this study relies on a small sample size.
- Direct costs sustained by the NHS were estimated based on elements collected through the questionnaires completed by caregivers and patients providing a partial estimation of the direct costs.
- SMA type III were excluded from the results of this study considering that only 2 patients were treated.

CONCLUSION

This preliminary analysis shows that indirect costs for patients treated with nusinersen tend to be lower compared to patients not treated [5] due to an increase of productivity for the caregivers.

REFERENCES

[1] SMA, A. F. from http://www.famigliesma.org/quante-comune-la-sma/
[2] C. Jones C, et al. (2015) Systematic review of incidence and prevalence of spinal muscular atrophy (SMA). "European Paediatric Neurology Society" (EPNS) tenutosi a Vienna,
27-30 maggio 2015. Poster PP09.1-2352
[3] Pharmastar - Octobre 2018: «SMA, la rivoluzione è cominciata»
[4] A. D'Amico, et al. (2011). " Spinal muscular atrophy."
[5] A. Marcellusi, et al. (2019). "Cost of illness of spinal muscular atrophy (SMA) in Italy".
[6] EPAR Spinraza

ACKNOWLEDGEMENTS

This study was sponsored by Biogen Italia Srl.

Note:NHS=National Health Service

Contact: andrea.marcellusi@uniroma2.it

Poster #**PRO30** Presented at 22th ISPOR Annual European Congress, Copenhagen, Denmark

CEIS Centre for Economic and International Studies TOR VERGATA Economic Evaluation and HTA *(EEHTA)*